APD588, a Novel, Selective S1P Receptor Modulator, Inhibits Inflammation and Airway Hyperresponsiveness in the Ovalbumin-induced Model of Allergic Asthma

Catherine M. Crosby,[1] H. Kiyomi Komori,[1] Ronald Christopher[2], John W. Adams[1]

[1]Arena Pharmaceuticals; [2]Parallel 33 Consulting

ABSTRACT

- **Background:** FTY720 is a non-selective sphingosine 1-phosphate (S1P) receptor modulator that has shown efficacy in preclinical models of asthma.[1,2] APD588 is a next-generation S1P receptor modulator with increased selectivity and potential for an improved safety profile. We hypothesized that the optimized S1P receptor profile of APD588 would confer beneficial effects in an ovalbumin (OVA)-induced asthma model.
- **Aim:** To evaluate efficacy of prophylactic APD588 in the OVA-induced allergic asthma model
- **Methods:** Receptor potency and selectivity profile of APD588 was determined (β-arrestin, GTPγS) in cell lines expressing recombinant human S1P receptors ($S1P_{1-5}$). Efficacy in allergic asthma was assessed in BALB/c mice which were sensitized with intraperitoneal OVA on days 0, 7, and 17, then exposed to intratracheal (IT) vehicle or OVA on days 21-23. From day 20, vehicle, APD588 or FTY720 were administered IT, QD. On day 24, airway hyperresponsiveness (AHR), immune cell infiltration and cytokine expression in BAL were measured.
- **Results:** APD588 was a potent modulator of $S1P_1$, with less potency at $S1P_{4,5}$, and no detectable activity at $S1P_{2,3}$. IT administration of APD588 produced a dose-dependent inhibition of methacholine-induced AHR, significantly inhibited airway eosinophil and lymphocyte infiltration, and inhibited BAL cytokines IL-4 and IL-5. FTY720 also reduced antigen-induced AHR and neutrophil and lymphocyte BAL cell counts, but had no significant effect on eosinophil counts or Th2 cytokines.
- **Conclusions:** APD588 was an effective IT therapy for OVA-induced asthma. Lung function improvements correlated with reduced Th2 cytokines and lymphocyte and eosinophil infiltration in the BAL. Based on these results, APD588 may have therapeutic potential in asthma, and broadly indicate potential in other atopic Th2-mediated diseases.

Presented at the *American Association of Immunologists* (AAI) Annual Meeting, San Diego; May 9–13 2019

METHODS

Figure 1: 588 Receptor Pharmacology

- APD588 is a potent modulator of $S1P_1$ while FTY720-p is a non-specific S1P receptor modulator. Receptor potency and selectivity profile of APD588 and FTY720-p were determined in cell lines expressing recombinant human S1P receptors ($S1P_{1-5}$). For each assay, values below indicate EC50, % efficacy of agonism (number of replicates).

APD588	$S1P_1$	$S1P_2$	$S1P_3$	$S1P_4$	$S1P_5$
β-Arrestin	0.64 nM, 116% (36)	>10,000 nM (21)	>10,000 nM (8)	123 nM, 32% (12)	12.2 nM, 66% (19)
GTPγS	4.4 nM, 85% (4)	>10,000 nM (2)	4.2 mM, 100% (3)	>10,000 nM (5)	397 nM, 75% (3)
FTY-720-p	**$S1P_1$**	**$S1P_2$**	**$S1P_3$**	**$S1P_4$**	**$S1P_5$**
β-Arrestin	2.95 nM, 111% (26)	>10,000 nM (15)	8.4 nM, 21% (7)	1.7 nM, 82% (13)	1.2 nM, 52% (17)
GTPγS	0.17 nM, 85% (2)	>10,000 nM (2)	3.7 nM, 100% (3)	1.3 nM, 27% (5)	0.67 nM, 55% (3)

Figure 2: Experimental Design

- Female Balb/c mice sensitized, treated, and challenged according to the schematic below. APD588 doses based on previous studies and potency. The 12.5 mg/kg dose for the comparator FTY720 was chosen based on previously published reports in this model.[1]

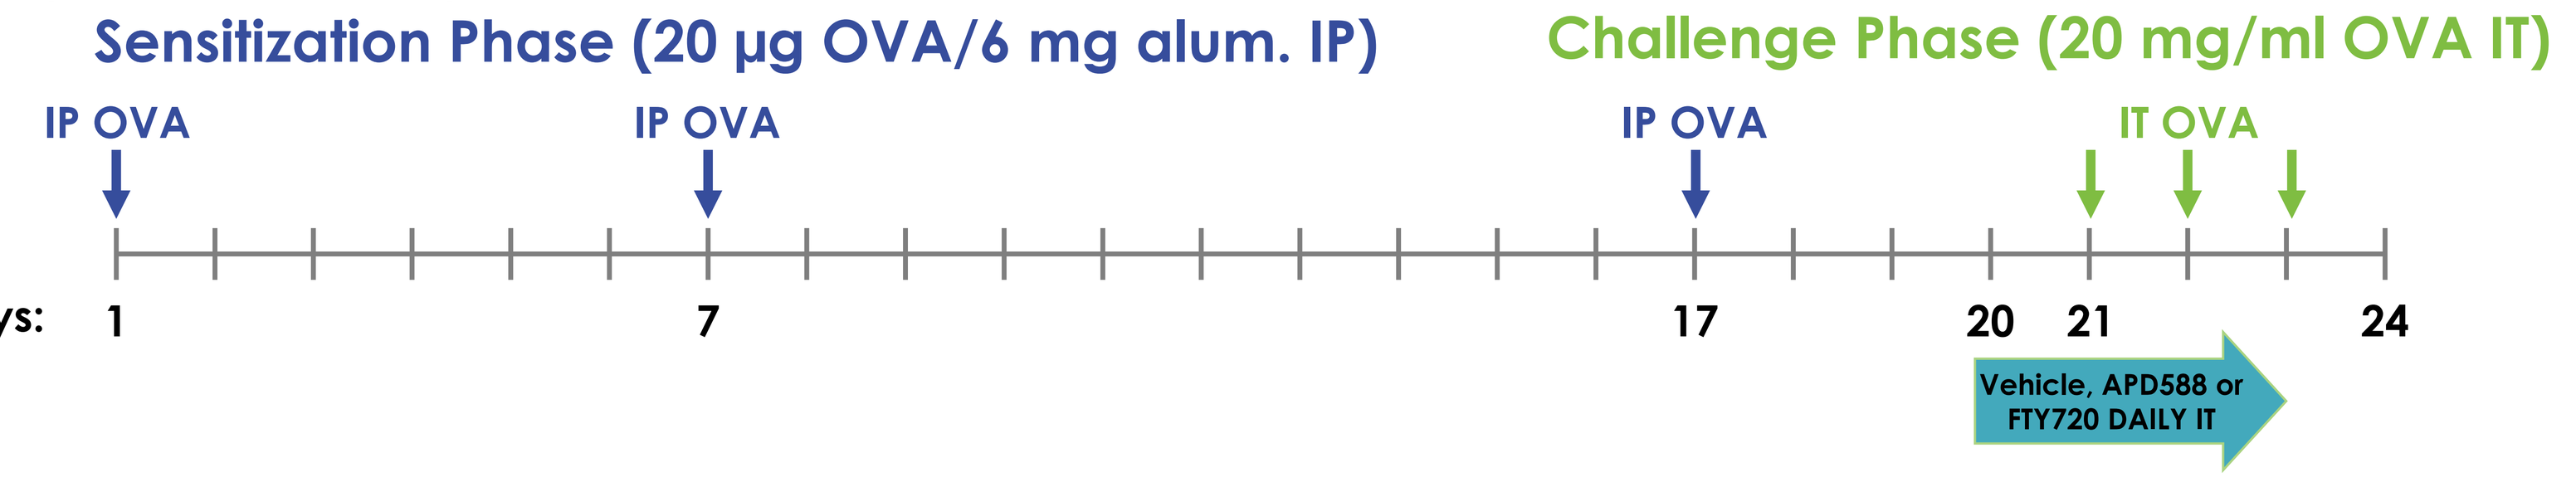
Sensitization Phase (20 µg OVA/6 mg alum. IP)
Challenge Phase (20 mg/ml OVA IT)
IP OVA
IP OVA
IP OVA
IT OVA
Days:
1
7
17
20
21
24
Vehicle, APD588 or FTY720 DAILY IT

Group	Sensitization Ova/Alum IP	Intratracheal treatment (*q.d.* 50 µL per mouse)	Dose (µg/kg)	Whole body challenge (30 min)	Endpoints
1	Yes	Vehicle	-	Saline	Cells, cytokines, AHR
2	Yes	Vehicle	-	Ova (20 mg/mL)	Cells, cytokines, AHR
3	Yes	APD588	12.5	Ova (20 mg/mL)	Cells, cytokines
4	Yes	APD588	37.5	Ova (20 mg/mL)	Cells, cytokines
5	Yes	APD588	112.5	Ova (20 mg/mL)	Cells, cytokines, AHR
6	Yes	FTY720	12.5	Ova (20 mg/mL)	Cells, cytokines, AHR

Figure 3: APD588 Reduces Lung Lymphocytes and Eosinophils

- Upon Ova challenge, IT-administered APD588 reduced the infiltration of (A) lymphocytes and (B) eosinophils in the BAL in a dose dependent manner.

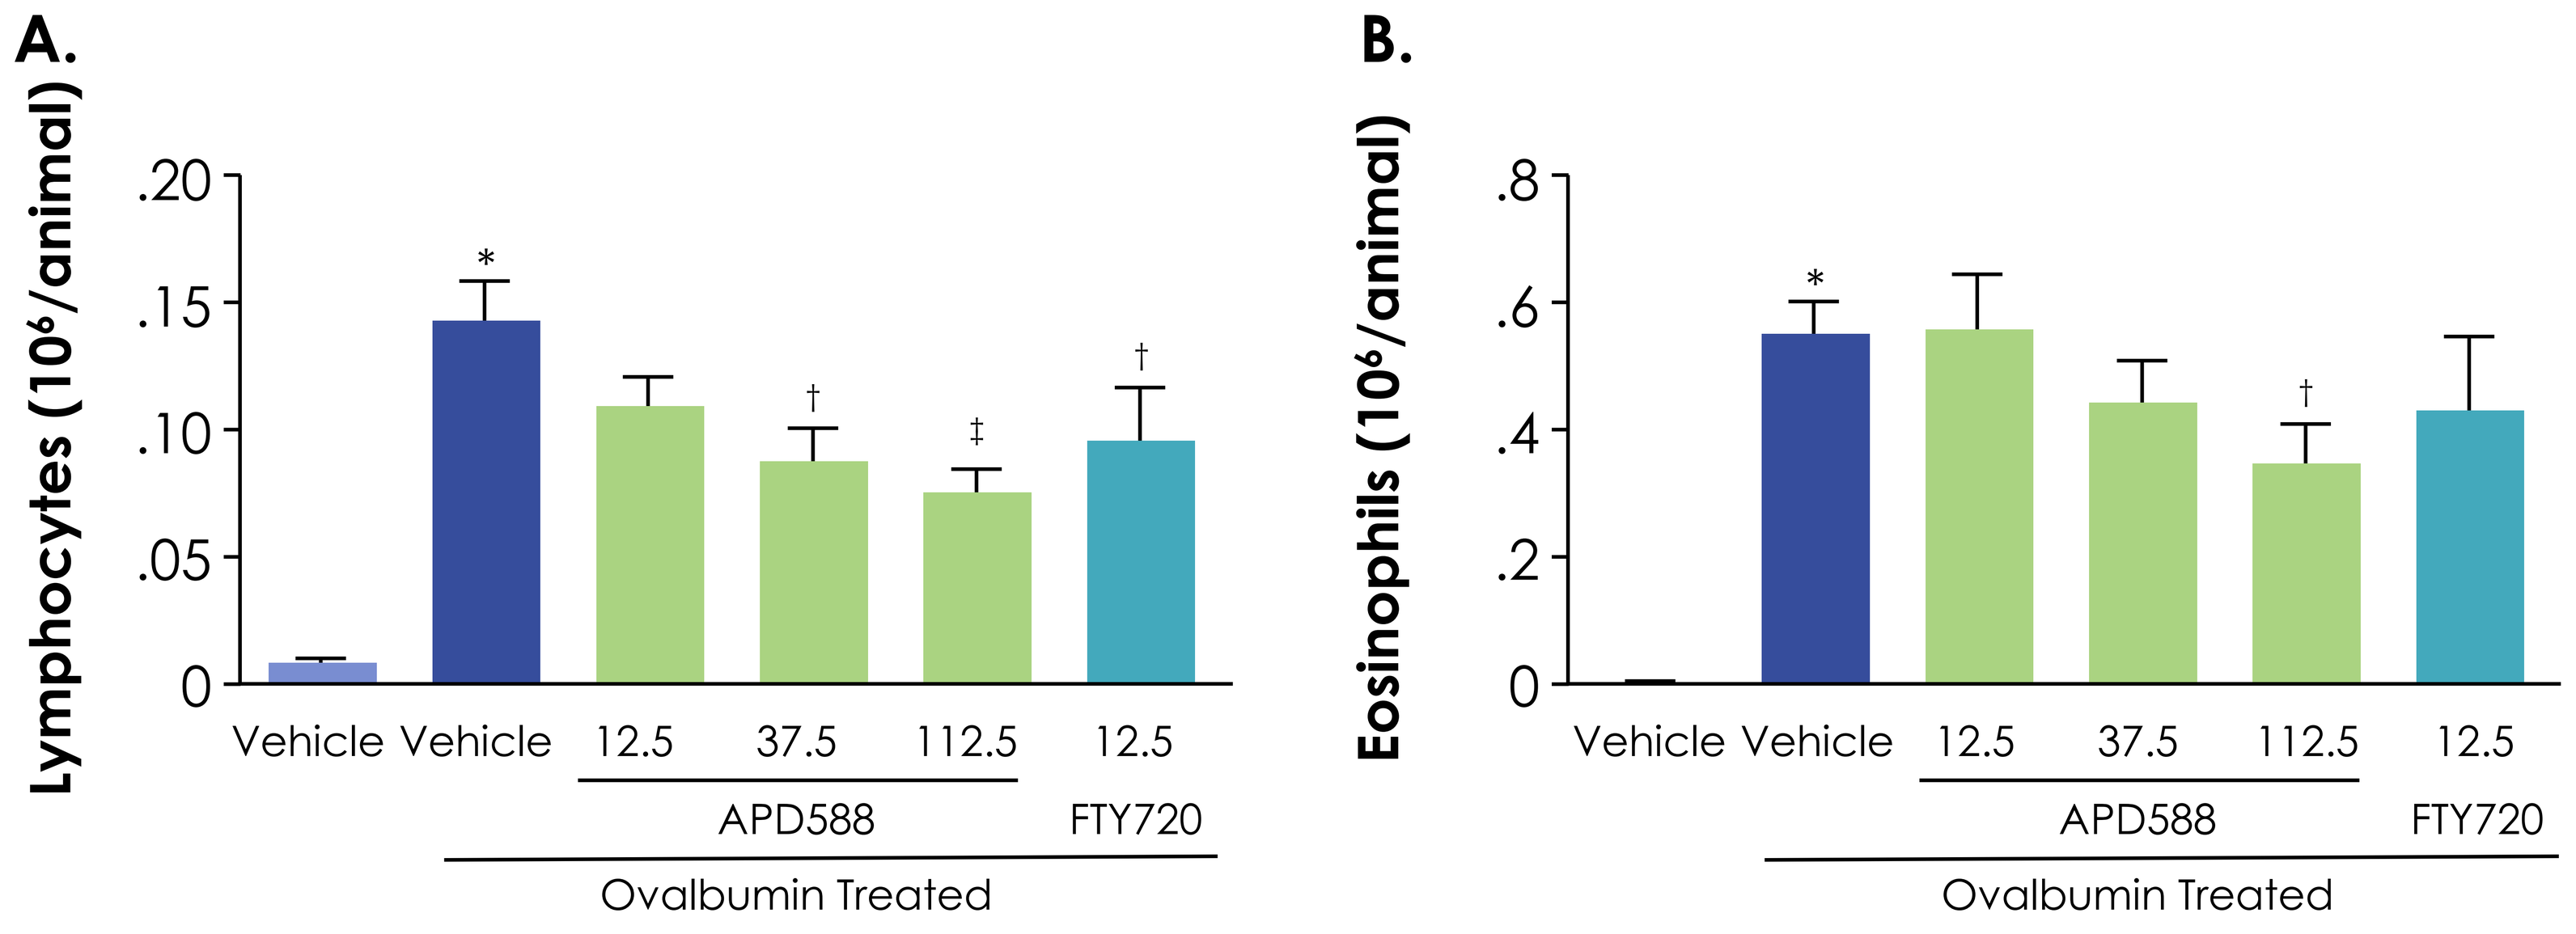
A.
Lymphocytes (10⁶/animal)
B.
Eosinophils (10⁶/animal)
Vehicle Vehicle 12.5 37.5 112.5 12.5
APD588
FTY720
Ovalbumin Treated

Data is shown as mean ± SEM. +*p<0.001 Vehicle vs Vehicle/Ova, †p<0.01 vs Vehicle/Ova, ‡p<0.001 vs Vehicle/Ova.

Figure 4: APD588 Reduces Th2 Cytokines in BAL

- APD588 reduced the concentration of (A) IL-4 and (B) IL-5 in the BAL in a dose dependent manner.

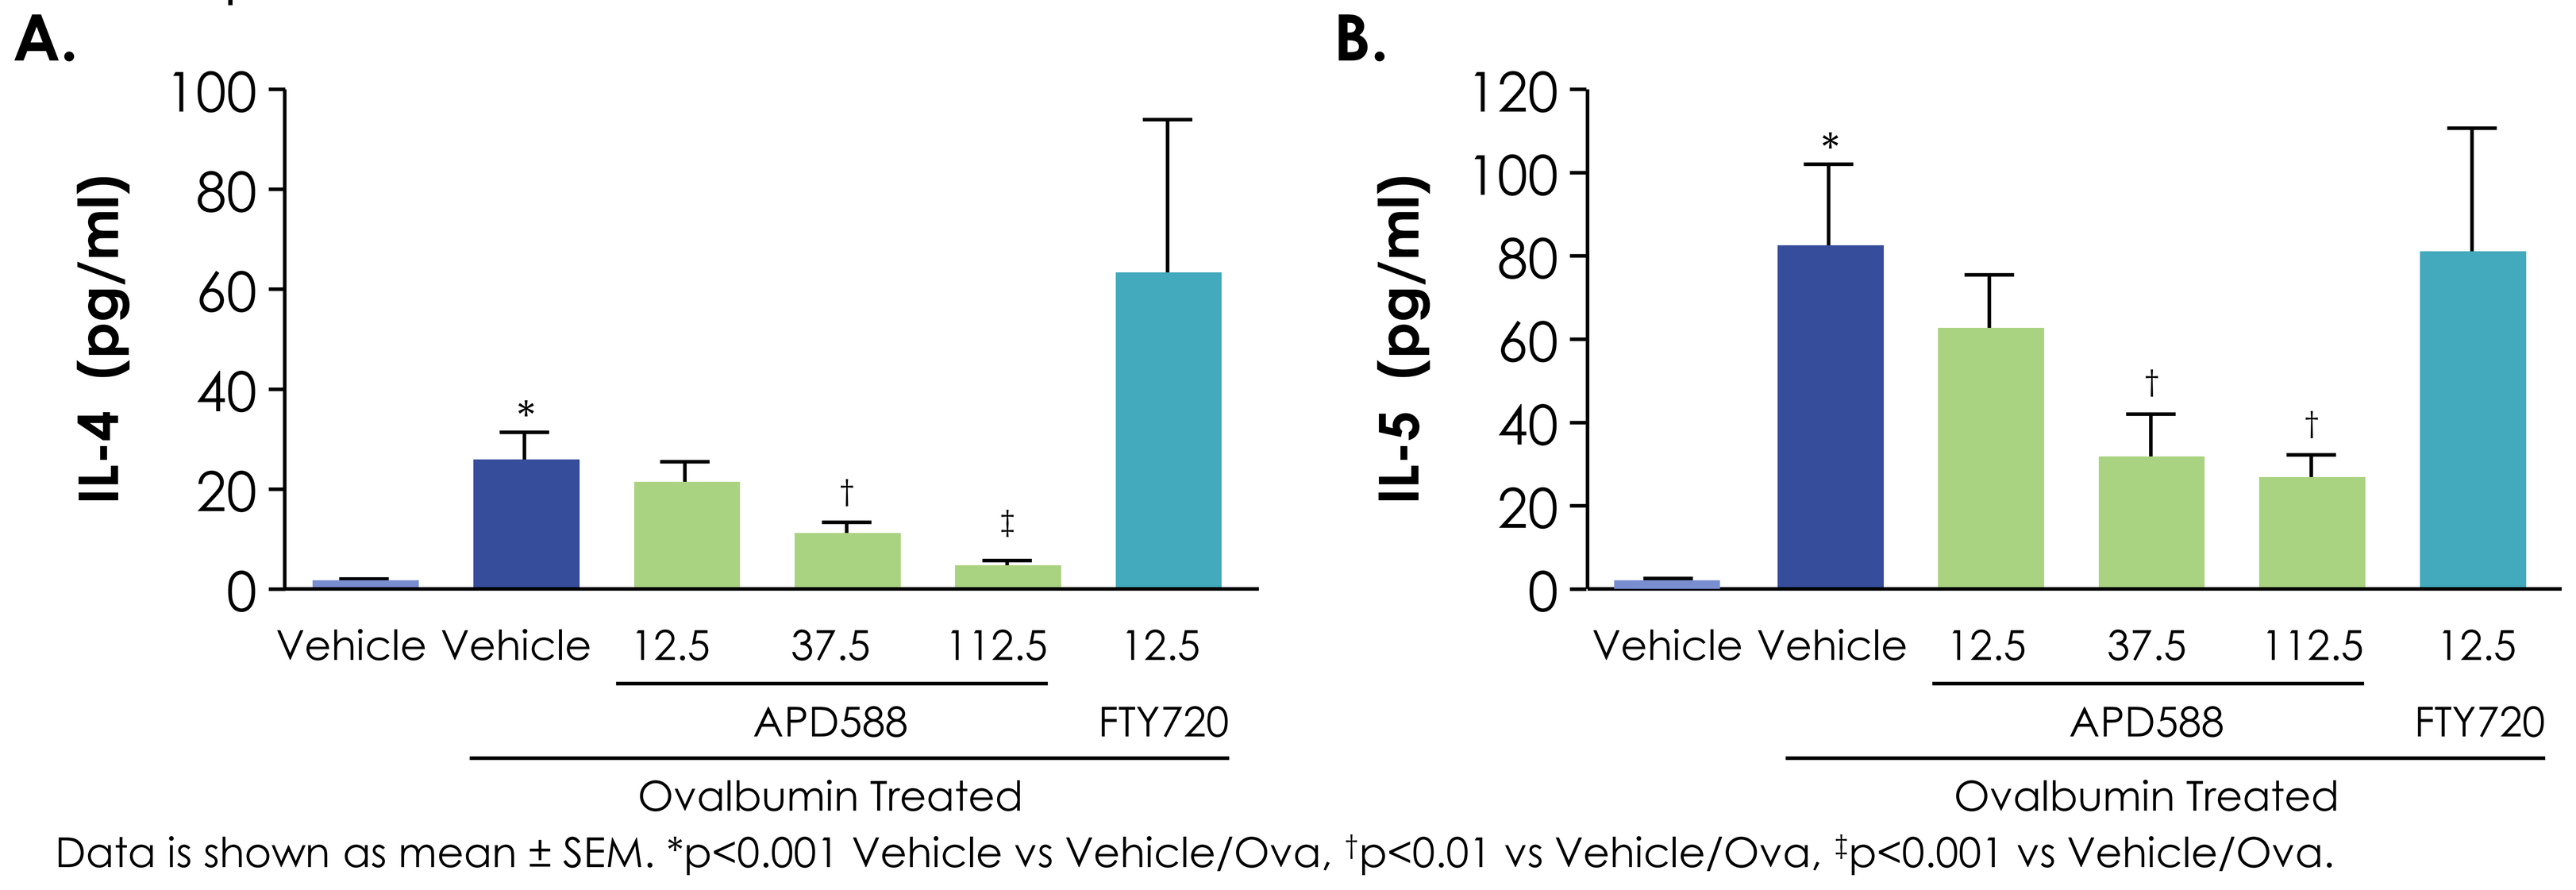
A.
IL-4 (pg/ml)
B.
IL-5 (pg/ml)
Vehicle Vehicle 12.5 37.5 112.5 12.5
APD588
FTY720
Ovalbumin Treated

Data is shown as mean ± SEM. *p<0.001 Vehicle vs Vehicle/Ova, †p<0.01 vs Vehicle/Ova, ‡p<0.001 vs Vehicle/Ova.

RESULTS

Figure 5: APD588 Inhibits Methacholine-induced Airway Hyperresponsiveness

APD588 inhibited airway responsiveness to methacholine (MCh), as measured by a reduction of (A,B) airway resistance and (C,D) airway compliance.

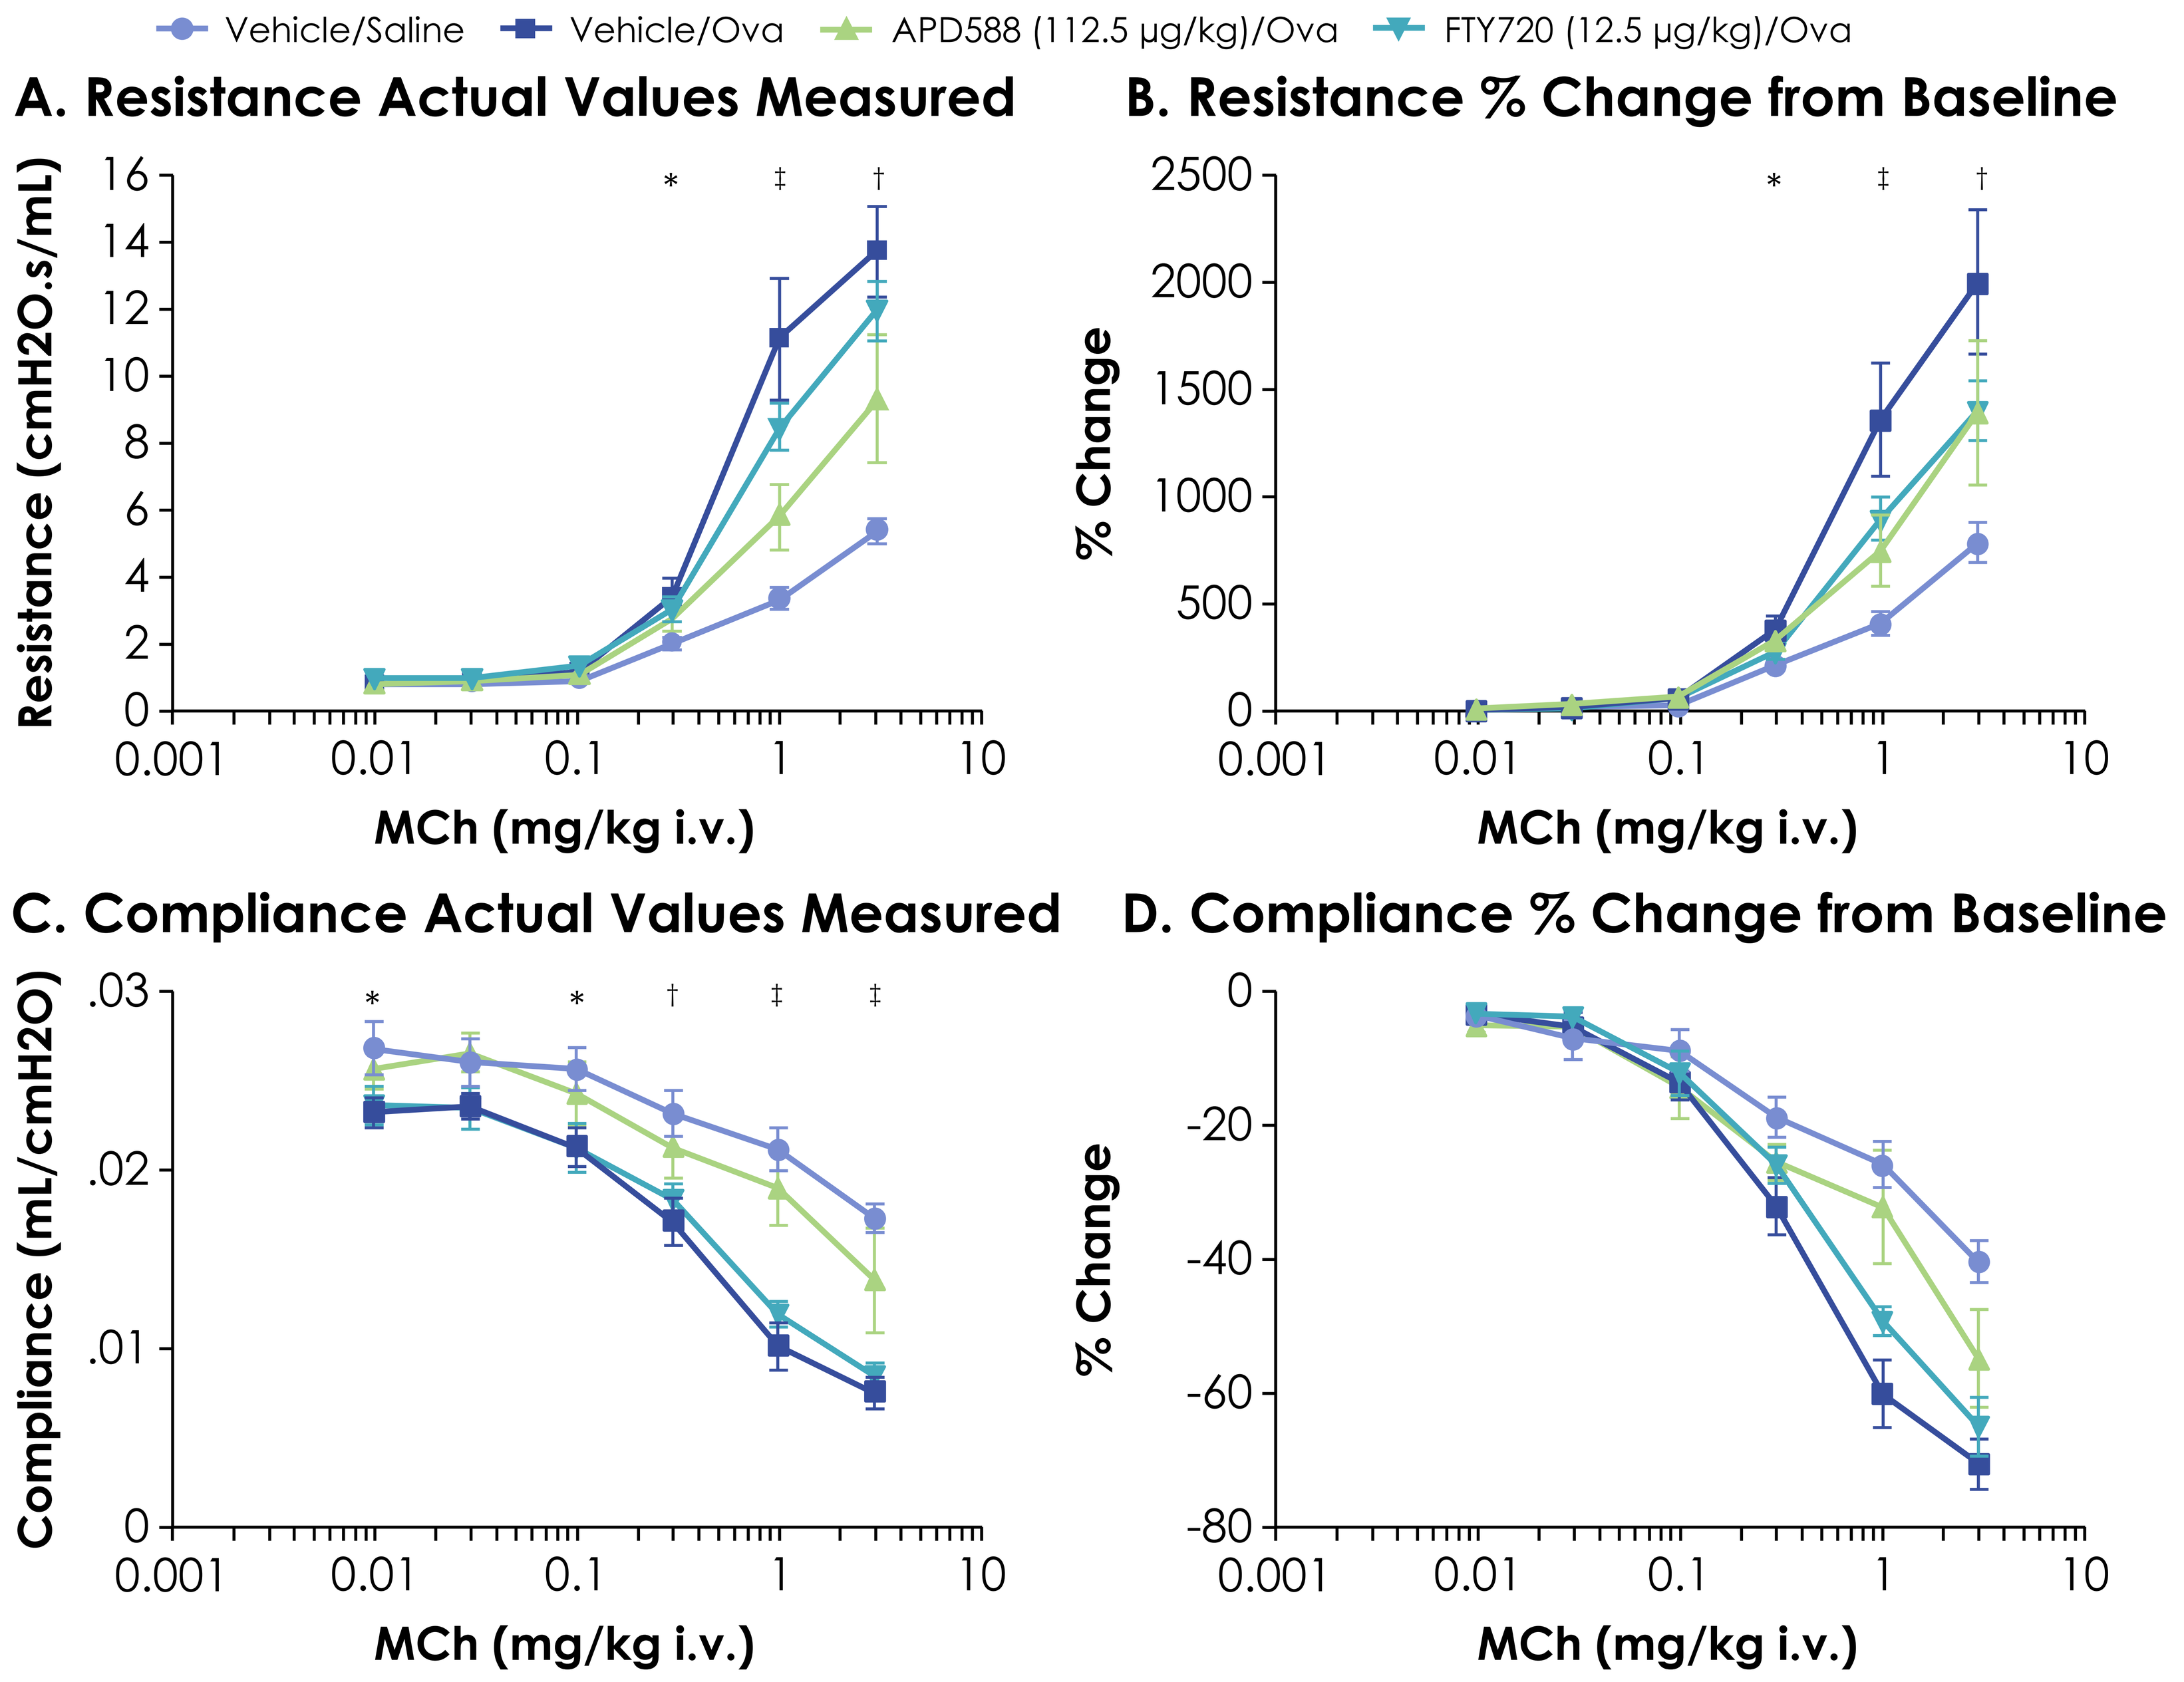
Vehicle/Saline
Vehicle/Ova
APD588 (112.5 µg/kg)/Ova
FTY720 (12.5 µg/kg)/Ova
A. Resistance Actual Values Measured
Resistance (cmH2O.s/mL)
B. Resistance % Change from Baseline
% Change
C. Compliance Actual Values Measured
Compliance (mL/cmH2O)
D. Compliance % Change from Baseline
% Change
MCh (mg/kg i.v.)

Data is shown as mean ± SEM. *p<0.05, †p<0.01, ‡p<0.001 APD588 vs Vehicle/Saline treated group.

CONCLUSIONS

- An aerosolized challenge of ovalbumin to the vehicle treated animals resulted in a significant increase in methacholine-induced airway hyperresponsiveness, increase in total and differential cell counts in the BAL fluid when compared to the vehicle treated saline challenged animals. Antigen challenge also induced increases in BAL pro-inflammatory cytokines, IL-4 and IL-5.
- In antigen challenged, sensitized mice, APD588 at 112.5 µg/kg significantly inhibited methacholine-induced airway hyperresponsiveness and significantly decreased eosinophil and lymphocyte counts. APD588 at 112.5 µg/kg also induced a statistically significant decrease in IL-4 and IL-5. APD588 dosed at 112.5 µg/kg also produced a significant reduction in the severity of the lung-induced tissue inflammatory response and mucous cell hyperplasia. APD588, when dosed at 37.5 µg/kg, produced a significant decrease in lymphocyte count, IL-4 and IL-5 in the BAL fluid. It also produced a significant reduction in the severity of the lung-induced inflammatory response in the tissue and mucous cell hyperplasia. FTY720 and APD588 dosed at the lowest concentration (12.5 µg/kg) did not show any statistically significant or noteworthy effects.
- In conclusion, APD588 was an effective IT therapy for OVA-induced asthma. Lung function improvements correlated with reduced Th2 cytokines and lymphocyte and eosinophil infiltration in the BAL. Notably, reductions in IL-5 correlated with reduced eosinophil recruitment. Based on these results, APD588 may have therapeutic potential in allergic and/or eosinophilic asthma subtypes, and broadly indicate potential in other atopic Th2-mediated diseases.

REFERENCES

1. Idzko et al. *J. Clin. Invest.*, 2006; 116 (11); 2935-44.
2. Sawicka et al. *J Immunol.*, 2003; 171 (11): 6206-14.

ACKNOWLEDGMENTS

These studies were supported by Arena Pharmaceuticals, Inc. (San Diego, CA). Sephirus Communications, Inc. (Woodland Hills, CA) provided medical writing support funded by Arena Pharmaceuticals, Inc.

DISCLOSURES

CC, KK, and JA are employees of Arena Pharmaceuticals, Inc.

ARENA PHARMACEUTICALS